ADDRESSING THE CHALLENGE OF CHRONICITY IN A REGION OF SPAIN. TAKING STEPS TOWARDS A PARTNERSHIP BETWEEN THE HEALTH CARE ADMINISTRATION AND THE PHARMACEUTICAL INDUSTRY

Sánchez-Agustino M.

Otsuka Pharmaceutical, S.A. (Santiago de Compostela-Spain)

OBJECTIVES: The growth of an ageing population and the subsequent rise in chronic conditions creates a worrisome trend in Spain. The proportion of the Spanish population aged 65 and over will reach 34.6% in 2066[1,2,3,4]. These changes are already exerting an overwhelming demand on health care. The objective of this project was for the Spanish Health Administration and the pharmaceutical industry to work together to identify new ways to ensure the quality and financial sustainability of health care, with a specific focus on adapting the model of care to better support chronic conditions.

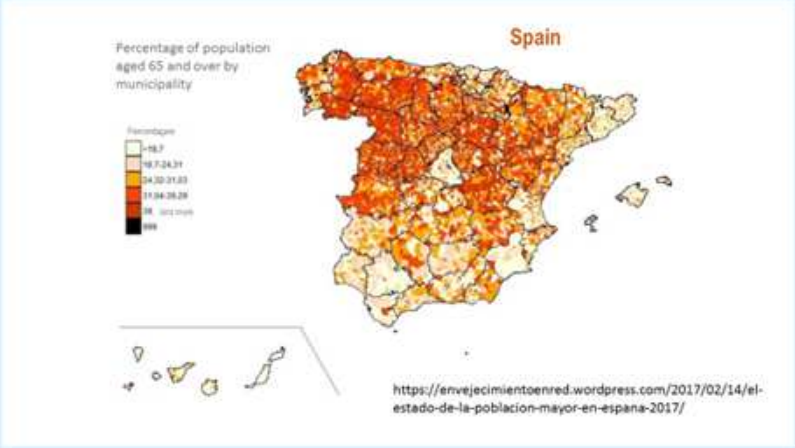
Spain
Percentage of population aged 65 and over by municipality
https://envejecimientoenred.wordpress.com/2017/02/14/el-estado-de-la-poblacion-mayor-en-espana-2017/

Source: 1 and 4

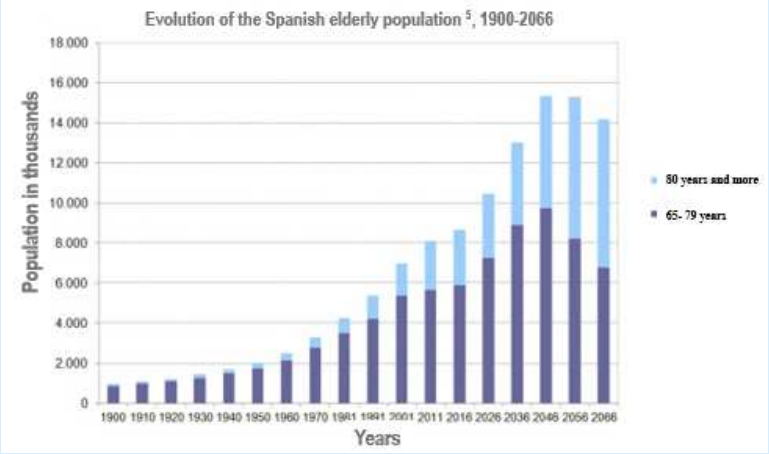
Evolution of the Spanish elderly population 5, 1900-2066
Population in thousands
18.000
16.000
14.000
12.000
10.000
8.000
6.000
4.000
2.000
0
1900 1910 1920 1930 1940 1950 1960 1970 1981 1991 2001 2011 2016 2026 2036 2046 2056 2066
Years
80 years and more
65- 79 years

Source: 4

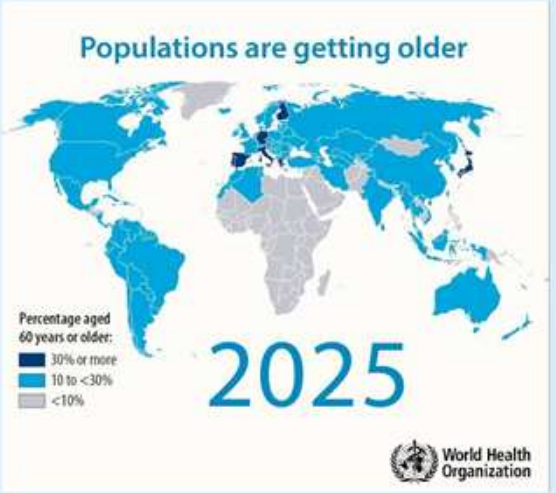
Populations are getting older
Percentage aged 60 years or older:
30% or more
10 to <30%
<10%
2025
World Health Organization

Source : WHO

METHODS: The project involved the organisation of several strategic reflection sessions with the aim of assisting the Administration in adopting a mid-term strategic focus and in understanding the importance of tackling what is important versus what is urgent. The participants made a commitment to act on the learning. We provide an example of this cooperation between the industry and regional administrations in Spain during several reflection sessions covering the following areas: creating a receptive context for chronic disease, increasing awareness for change, updating on international trends, partnership building, technology and health and leadership for implementation.

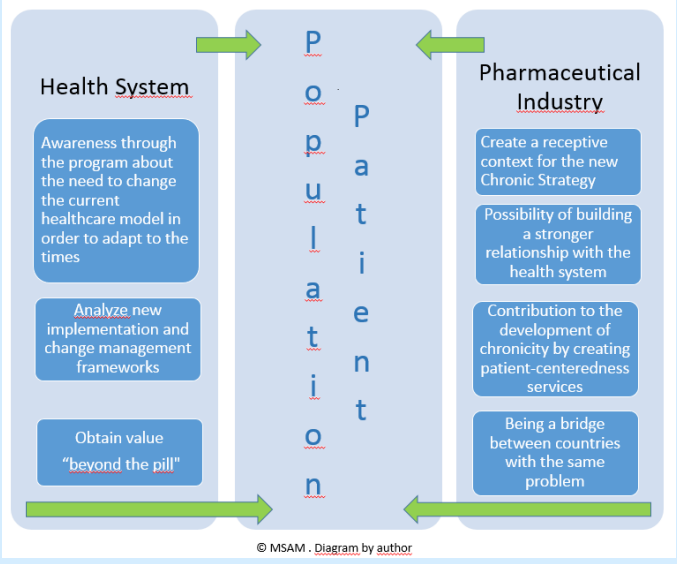
Health System
Awareness through the program about the need to change the current healthcare model in order to adapt to the times
Analyze new implementation and change management frameworks
Obtain value "beyond the pill"
Population
Patient
Pharmaceutical Industry
Create a receptive context for the new Chronic Strategy
Possibility of building a stronger relationship with the health system
Contribution to the development of chronicity by creating patient-centeredness services
Being a bridge between countries with the same problem
© MSAM . Diagram by author

By M. Sanchez-Agustino

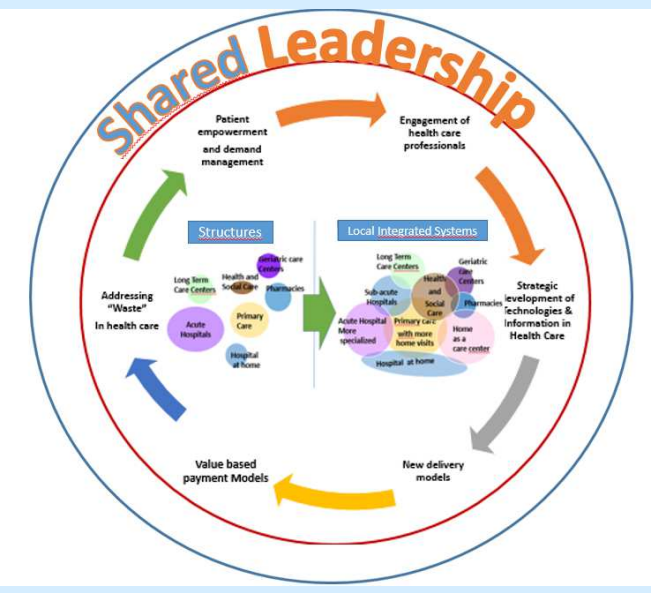
Shared Leadership
Patient empowerment and demand management
Engagement of health care professionals
Structures
Local Integrated Systems
Addressing "Waste" in health care
Strategic development of technologies & information in Health Care
Value based payment Models
New delivery models

Adapted by author from Policy Streams for Health care Transformation. 2017

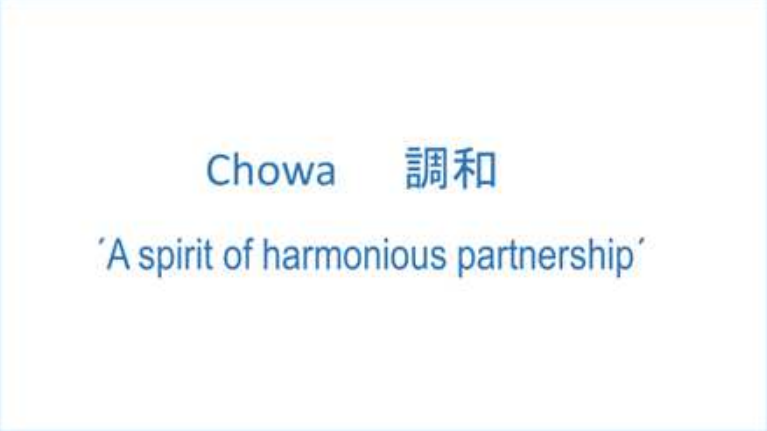
Chowa 調和
´A spirit of harmonious partnership´

By M. Sánchez-Agustino

***RESULTS*:** These reflection sessions have provided new opportunities for the industry to potentially form strategic value-based[5] public-private partnerships with the Administration of Regions in Spain.

CONCLUSIONS: This project has provided an opportunity for the pharmaceutical industry, to go "beyond the pill", to support government efforts in transforming health care to meet the chronicity challenge. The Administration was introduced to and can therefore use new frameworks such as the Triple Aim[6] (population health, better care and lower costs) and chronic disease strategies. Certain Spanish regions could potentially become a reference in the management of chronicity as a prototype to be followed by other Spanish communities together with other European countries.

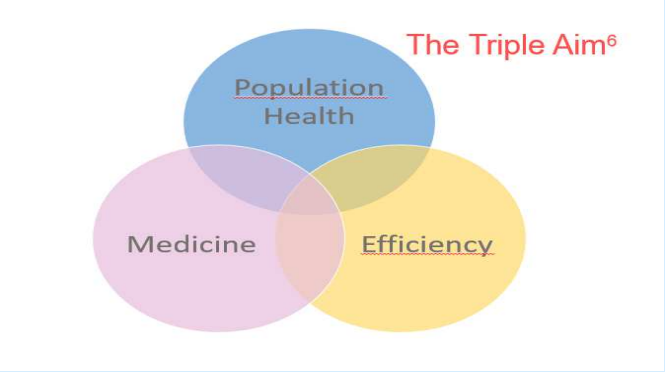
The Triple Aim6
Population Health
Medicine
Efficiency

Source: 6

REFERENCES:

1. Instituto Nacional de estadística. Proyecciones de Población 2016–2066. Octubre de 2016. Ministerio de Economía y Hacienda. España. http://www.ine.es/prensa/np994.pdf
2. Cifras de población del Instituto Gallego de Estadística. Xunta de Galicia. España https://www.ige.eu/web/index.jsp?paxina=001&idioma=gl
3. Percentage of the world population Age 65 + in 2015 and 2050. United States Census Bureau. https://www.census.gov/newsroom/press-releases/2016/cb16-54.html
4. Spanish aging. Envejecimiento en España. Ministerio de Economía y competitividad. Consejo Superior de investigaciones científicas (CSIC) https://envejecimientoenred.wordpress.com/2017/02/14/el-estado-de-la-poblacion-mayor-en-espana-2017/
5. Sánchez-Agustino M. El nuevo relato transformador de la sanidad: impacto sobre el sector farmacéutico. Pharmacoecon Span Res Artic (2017) 14:27-30 DOI 10.2017/s40277. rdcu.be/o09O
6. Bengoa R, Arratíbel P. Implementando la triple meta: " lecciones" desde el ámbito directivo. Papeles de Economía Española. 2014;(142):7-13.

ISPOR Europe 2018. 10-14 November 2018. Barcelona-Spain